ART Initiation and Retention in After-hour versus Daily Male Health Clinics

Tali Cassidy[1], Amir Shroufi[2], Sarah Jane Steele[2], Morna Cornell[3], Virginia de Azevedo[4], David Binza[5], Rodd Gerstenhaber[2]

[1]Médecins Sans Frontières, Khayelitsha, Cape Town, South Africa; [2]Médecins Sans Frontières, Mowbray, Cape Town, South Africa;
[3]Centre for Infectious Disease Epidemiology & Research, School of Public Health & Family Medicine,
University of Cape Town; [4]City of Cape Town Department of Health, Khayelitsha, Cape Town, South Africa;
[5]Western Cape Provincial Department of Health, Cape Town, South Africa

Contact: Tali Cassidy
msfocb-khayelitsha-epid@brussels.msf.org

Introduction

- In large African programs, disproportionately fewer men initiate ART, and they do so at a more advanced disease stage.
- Since 2014 MSF, the City of Cape Town and the Western Cape Provincial Department of Health, run two male services in Site B: a day clinic and an after-hours clinic
- Setting: Khayelitsha, a high HIV-prevalence township, Cape Town

Methods

- Data collected from routine records, June 2014 -September 2016
- Retention defined as the % remaining in care, excluding those transferring to another clinic
- Transfer-ins initiated elsewhere and transferred to clinic
- Eligibility criteria defined by South African national guidelines
- Ethical approval was obtained from the University of Cape Town.

Description of Services

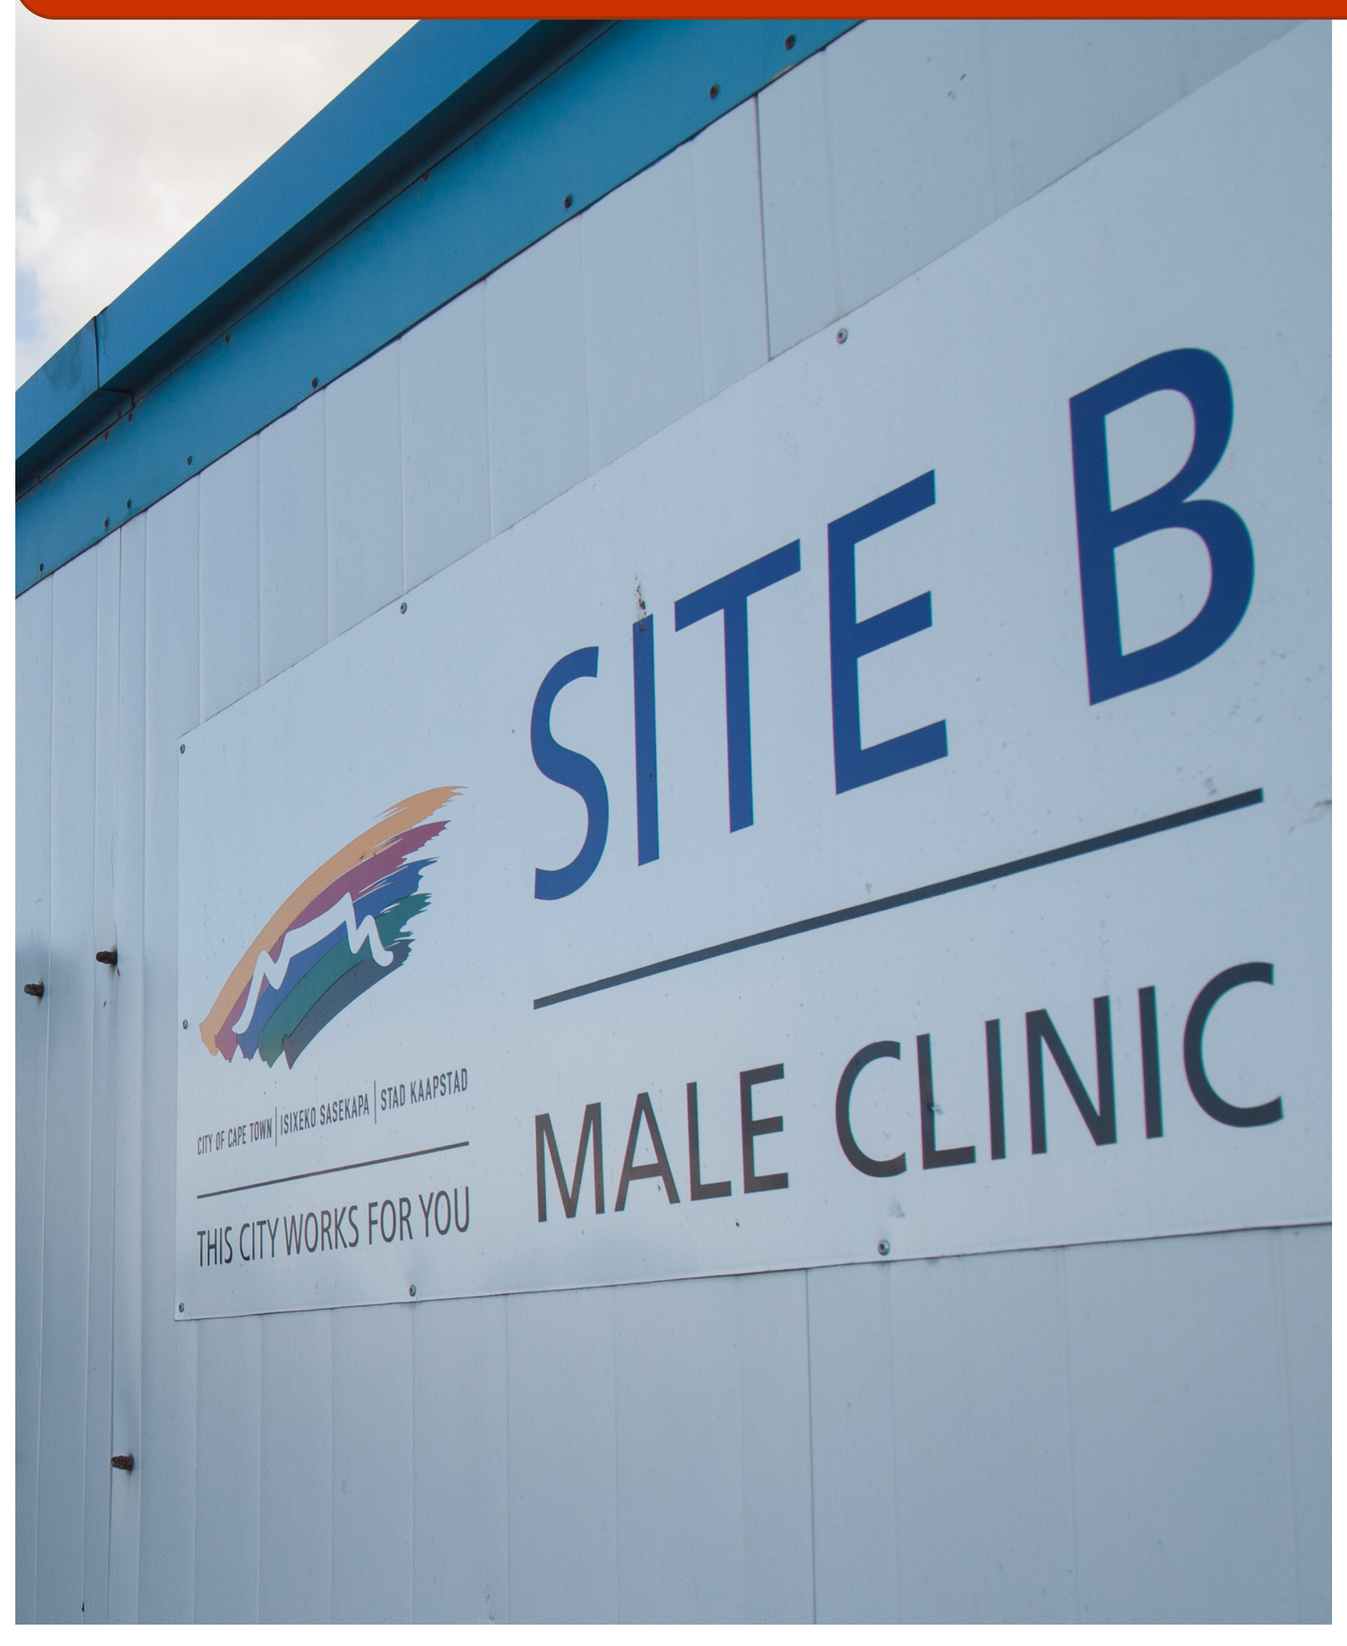

SITE B
MALE CLINIC
THIS CITY WORKS FOR YOU

Male day clinic

Monday – Friday
8:00-16:30
Stand-alone clinic

Both

All-male staff
STI diagnosis & treatment
HIV testing & counseling
Point-of-care CD4 count
Initiate and dispense ART
ART adherence clubs

Male After-hours

Wednesdays
16:00 – 19:30
On premises of larger facility

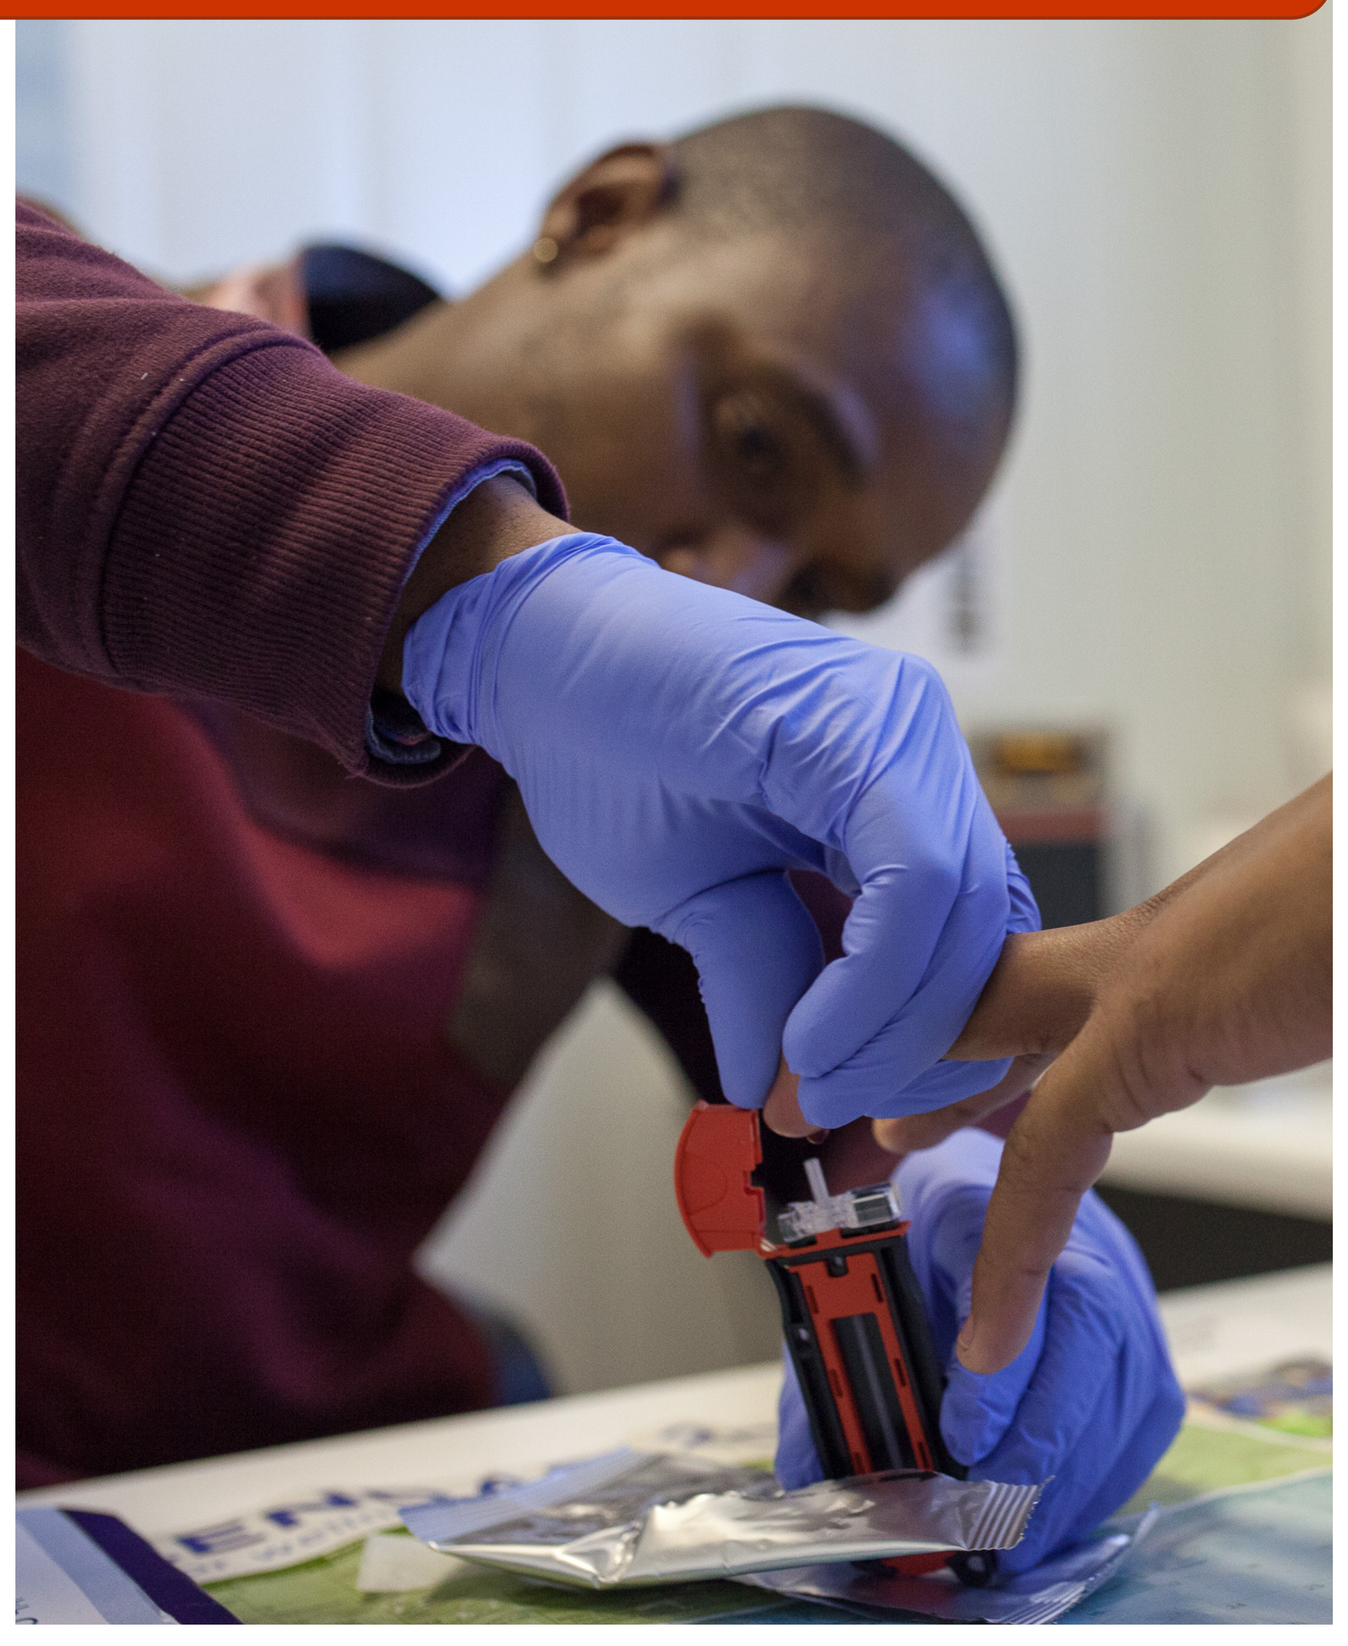

Results

Clinic Characteristics	Day Clinic	After-hours
Median visits per month (IQR)	**529 (475-579)**	75 (21-110)
% of visits addressing STIs	**45%**	21%
Median HCT/month (IQR)	291 (238-319)	25 (10-35)
% positive	5.9%	**8.5%**
Median CD4 count at test (IQR)	**384 (268-511)**	330 (241-453)
ART Initiation		
% of eligible initiating	70%	**91%**
Median Age at initiation (IQR)	31.8 (27.6-38.2)	**34 (29.5-38.5)**
Total initiated by end 2015	210	52
% retained at 6 months	88%	**98%**
Transfer-ins		
% of cohort transferred in	8%	**43%**
Median years on ART at transfer	1.9	3.5
Transferred in by end 2015	22	61
% retained at 6 months	91%	89%

Summary

Day clinic has higher:
- monthly visits
- % STI-related visits (p<0.001)
- CD4 count at testing (p=0.14)

After-hours clinic has higher:
- HIV positivity (p=0.004)
- rate of ART initiation (p<0.001)
- age at initiation (0.004)
- Retention among initiates (p=0.02)
- % transfer-ins (p<0.001)

Conclusions

- While the day clinic had more, and younger, clients, the after-hours clinic attracted a different population, and had higher initiation and retention rates. STI care is an excellent opportunity to link men to HIV services.
- Given these contrasting successes, further research should investigate whether aspects of both services could be rolled out to attract more men to HIV services.

Poster #998