PRESENTATION NO: 718

DOES TIME TO ART INITIATION IMPACT EARLY RESPONSES TO DR-TB TREATMENT?

Mohr E[1], Daniels J[1], Muller O[1], Chabalala B[1], HughesJ[1], De Azevedo V[2], Shroufi A[3], Steele SJ[3], Ferlazzo G[4], Trivino Duran L[1]

[1]Médecins Sans Frontières (MSF), Khayelitsha
[2]Cape Town City Health Department
[3]Médecins Sans Frontières (MSF), Cape Town
[4]Médecins Sans Frontières (MSF), South African Medical Unit, Cape Town

BACKGROUND

Morbidity and mortality among DR-TB patients co-infected with HIV can be reduced by anti-retroviral treatment (ART). There is limited evidence on the best time to initiate ART following DR-TB treatment initiation.

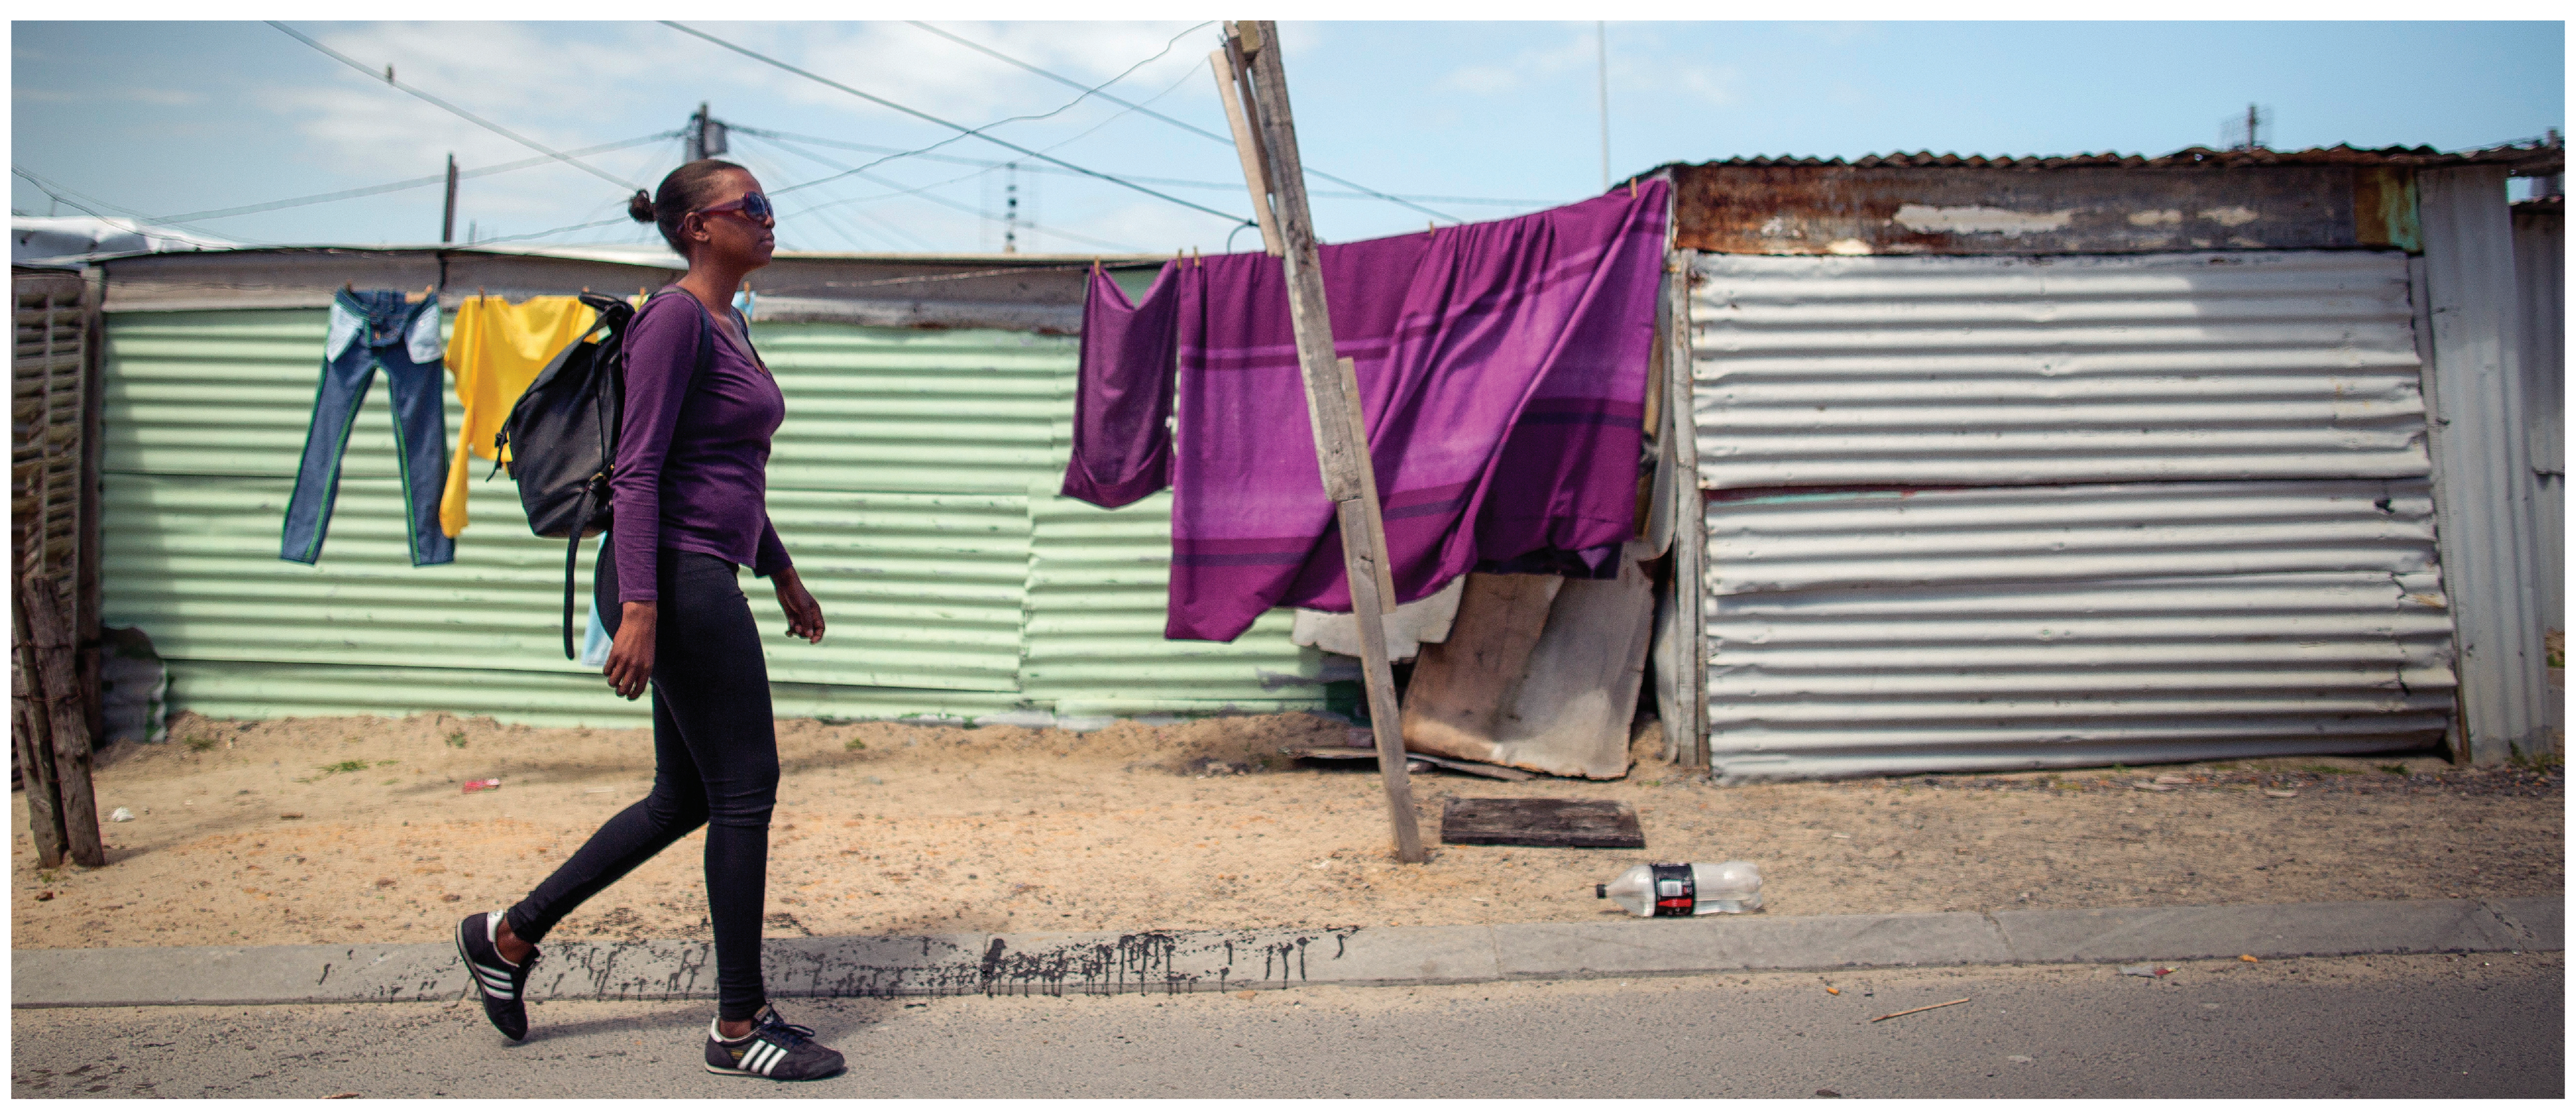

The peri-urban township of Khayelitsha, South Africa

METHODS

We conducted a retrospective analysis of pulmonary DR-TB patients, co-infected with HIV who initiated treatment in Khayelitsha, South Africa, January 2014-December 2015. Culture conversion and mortality following 6-months of DR-TB treatment, were stratified by time of ART initiation and univatiate logistic regression analyses were used to calculate odds ratios, 95% confidence intervals and p-values.

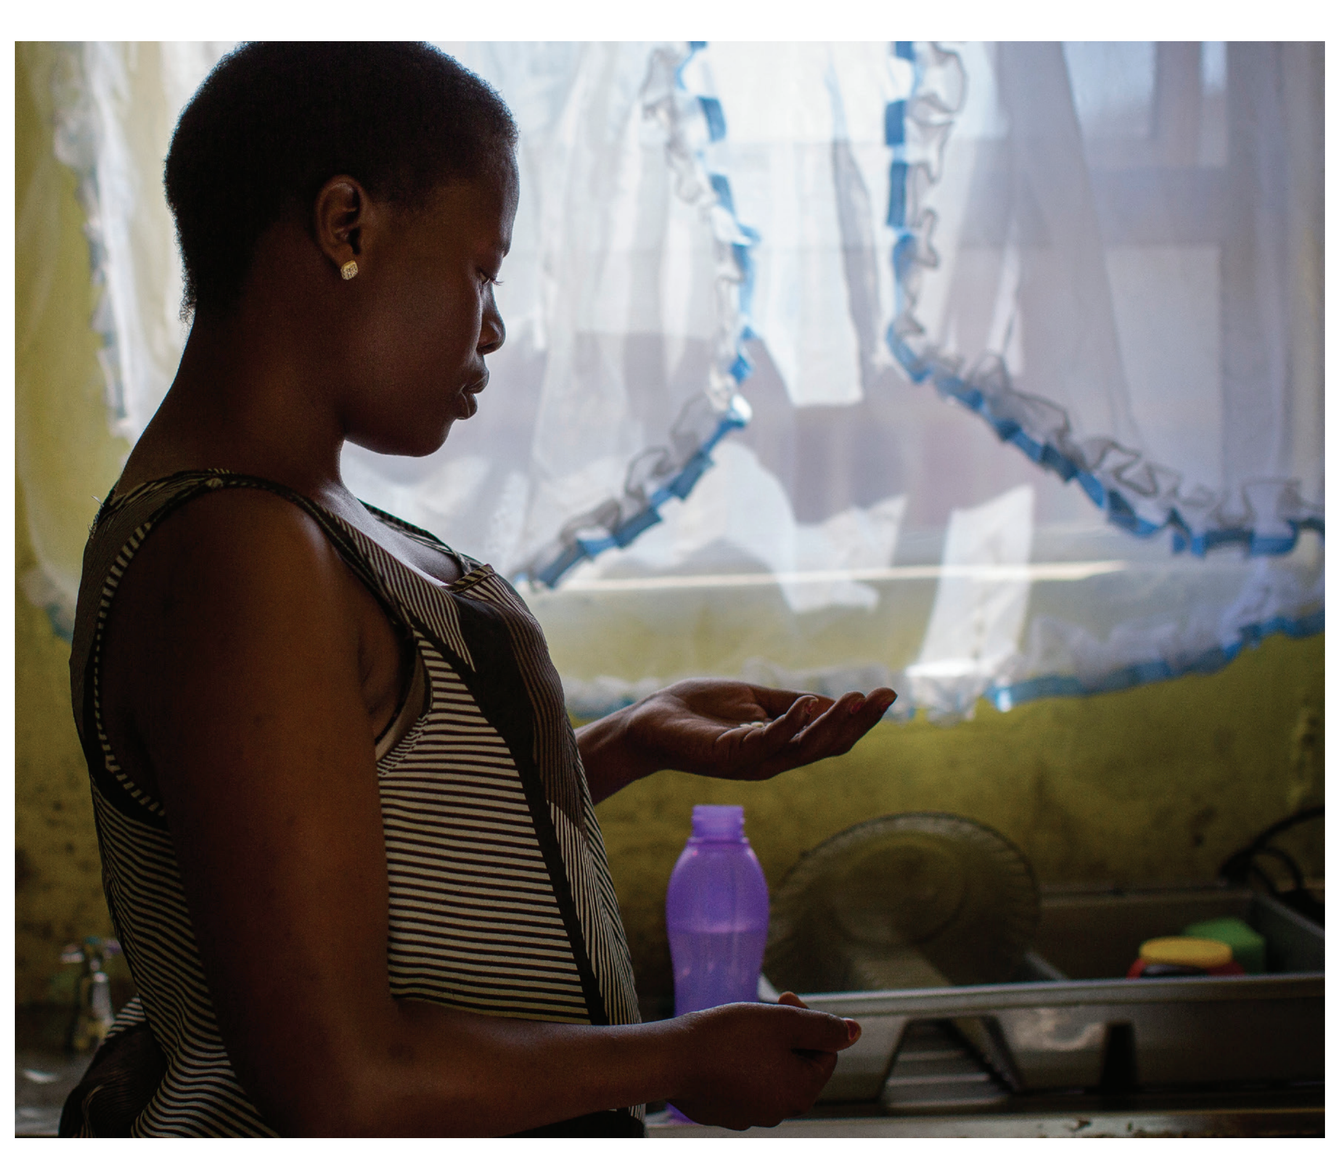

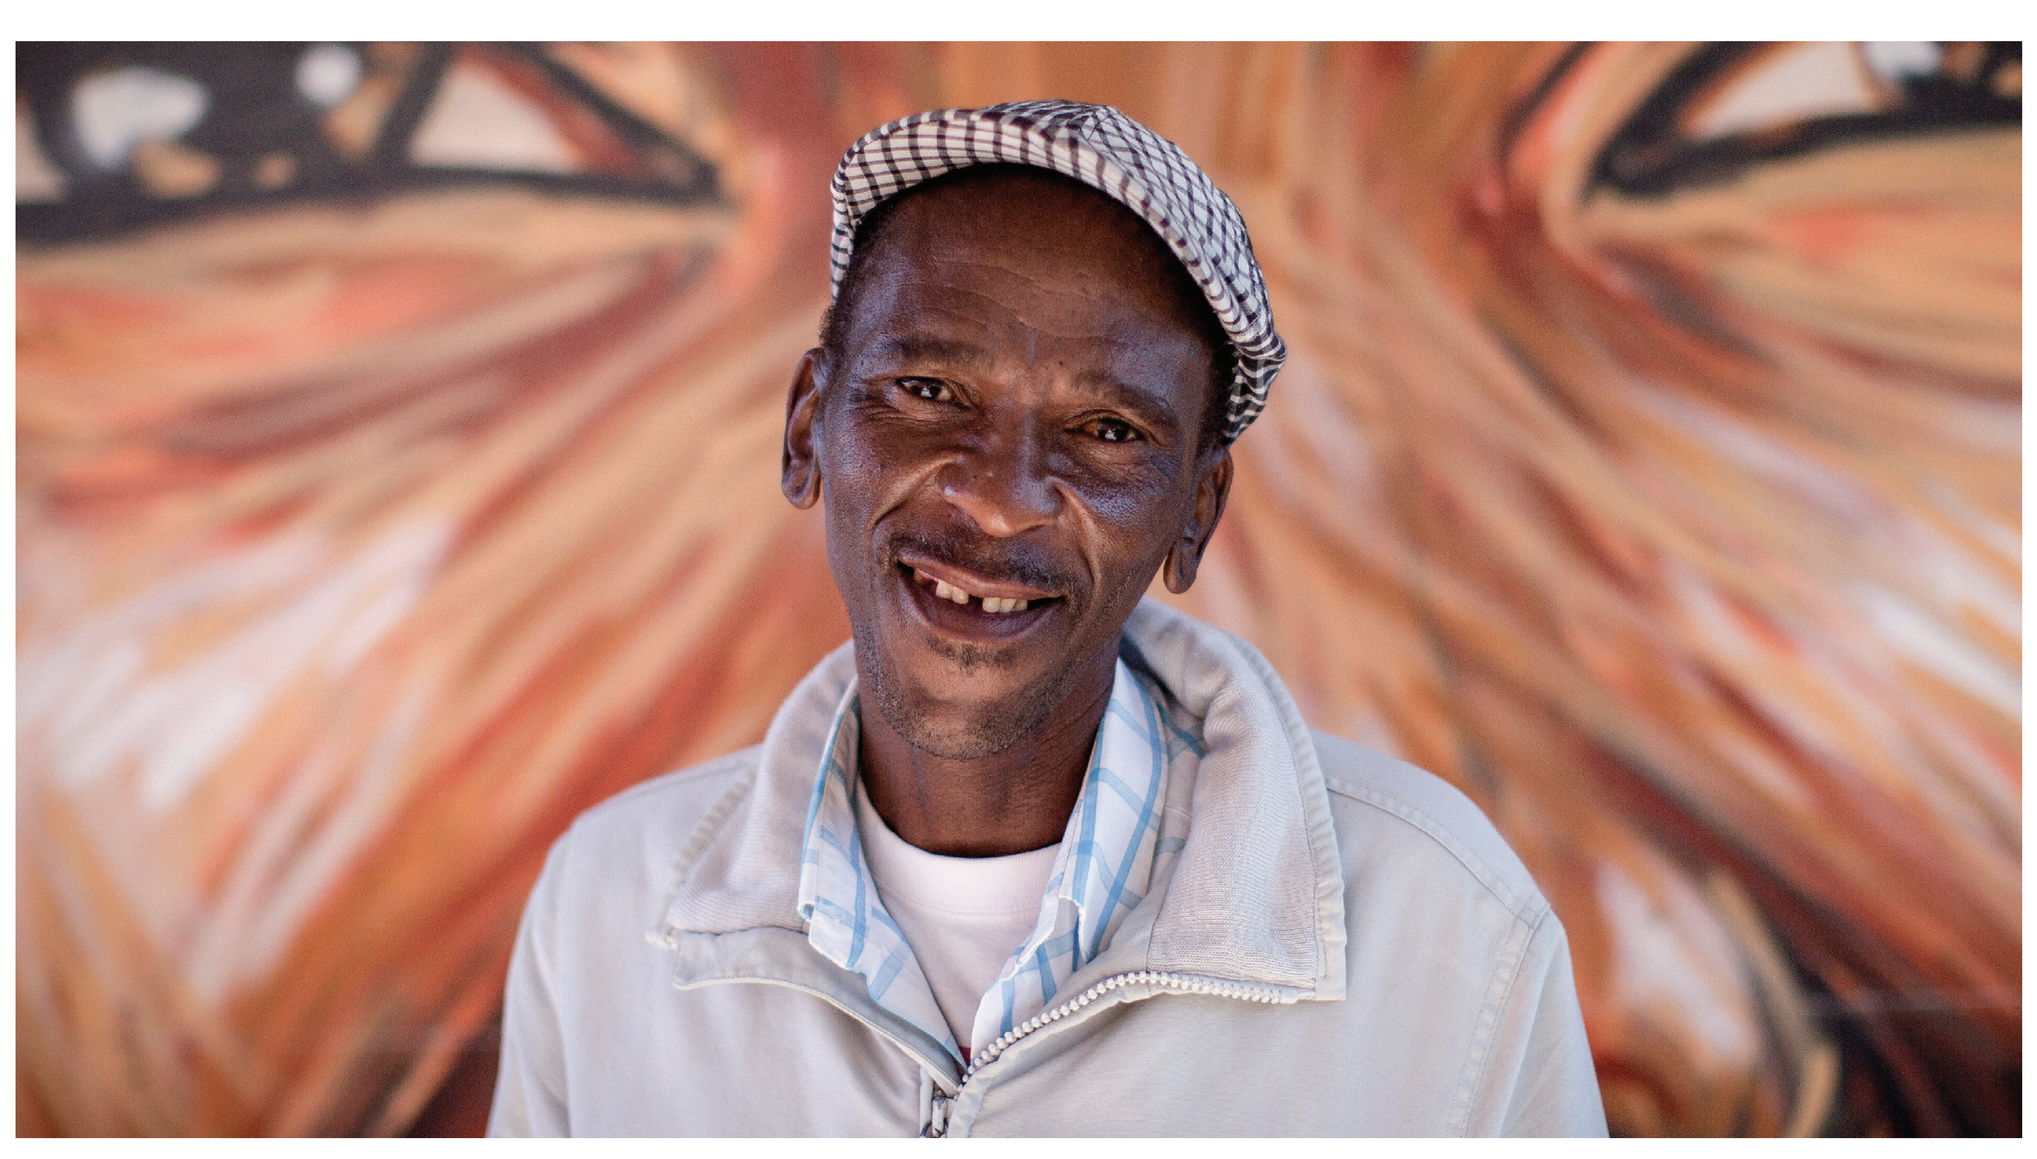

DR-TB patients from Khayelitsha South Africa

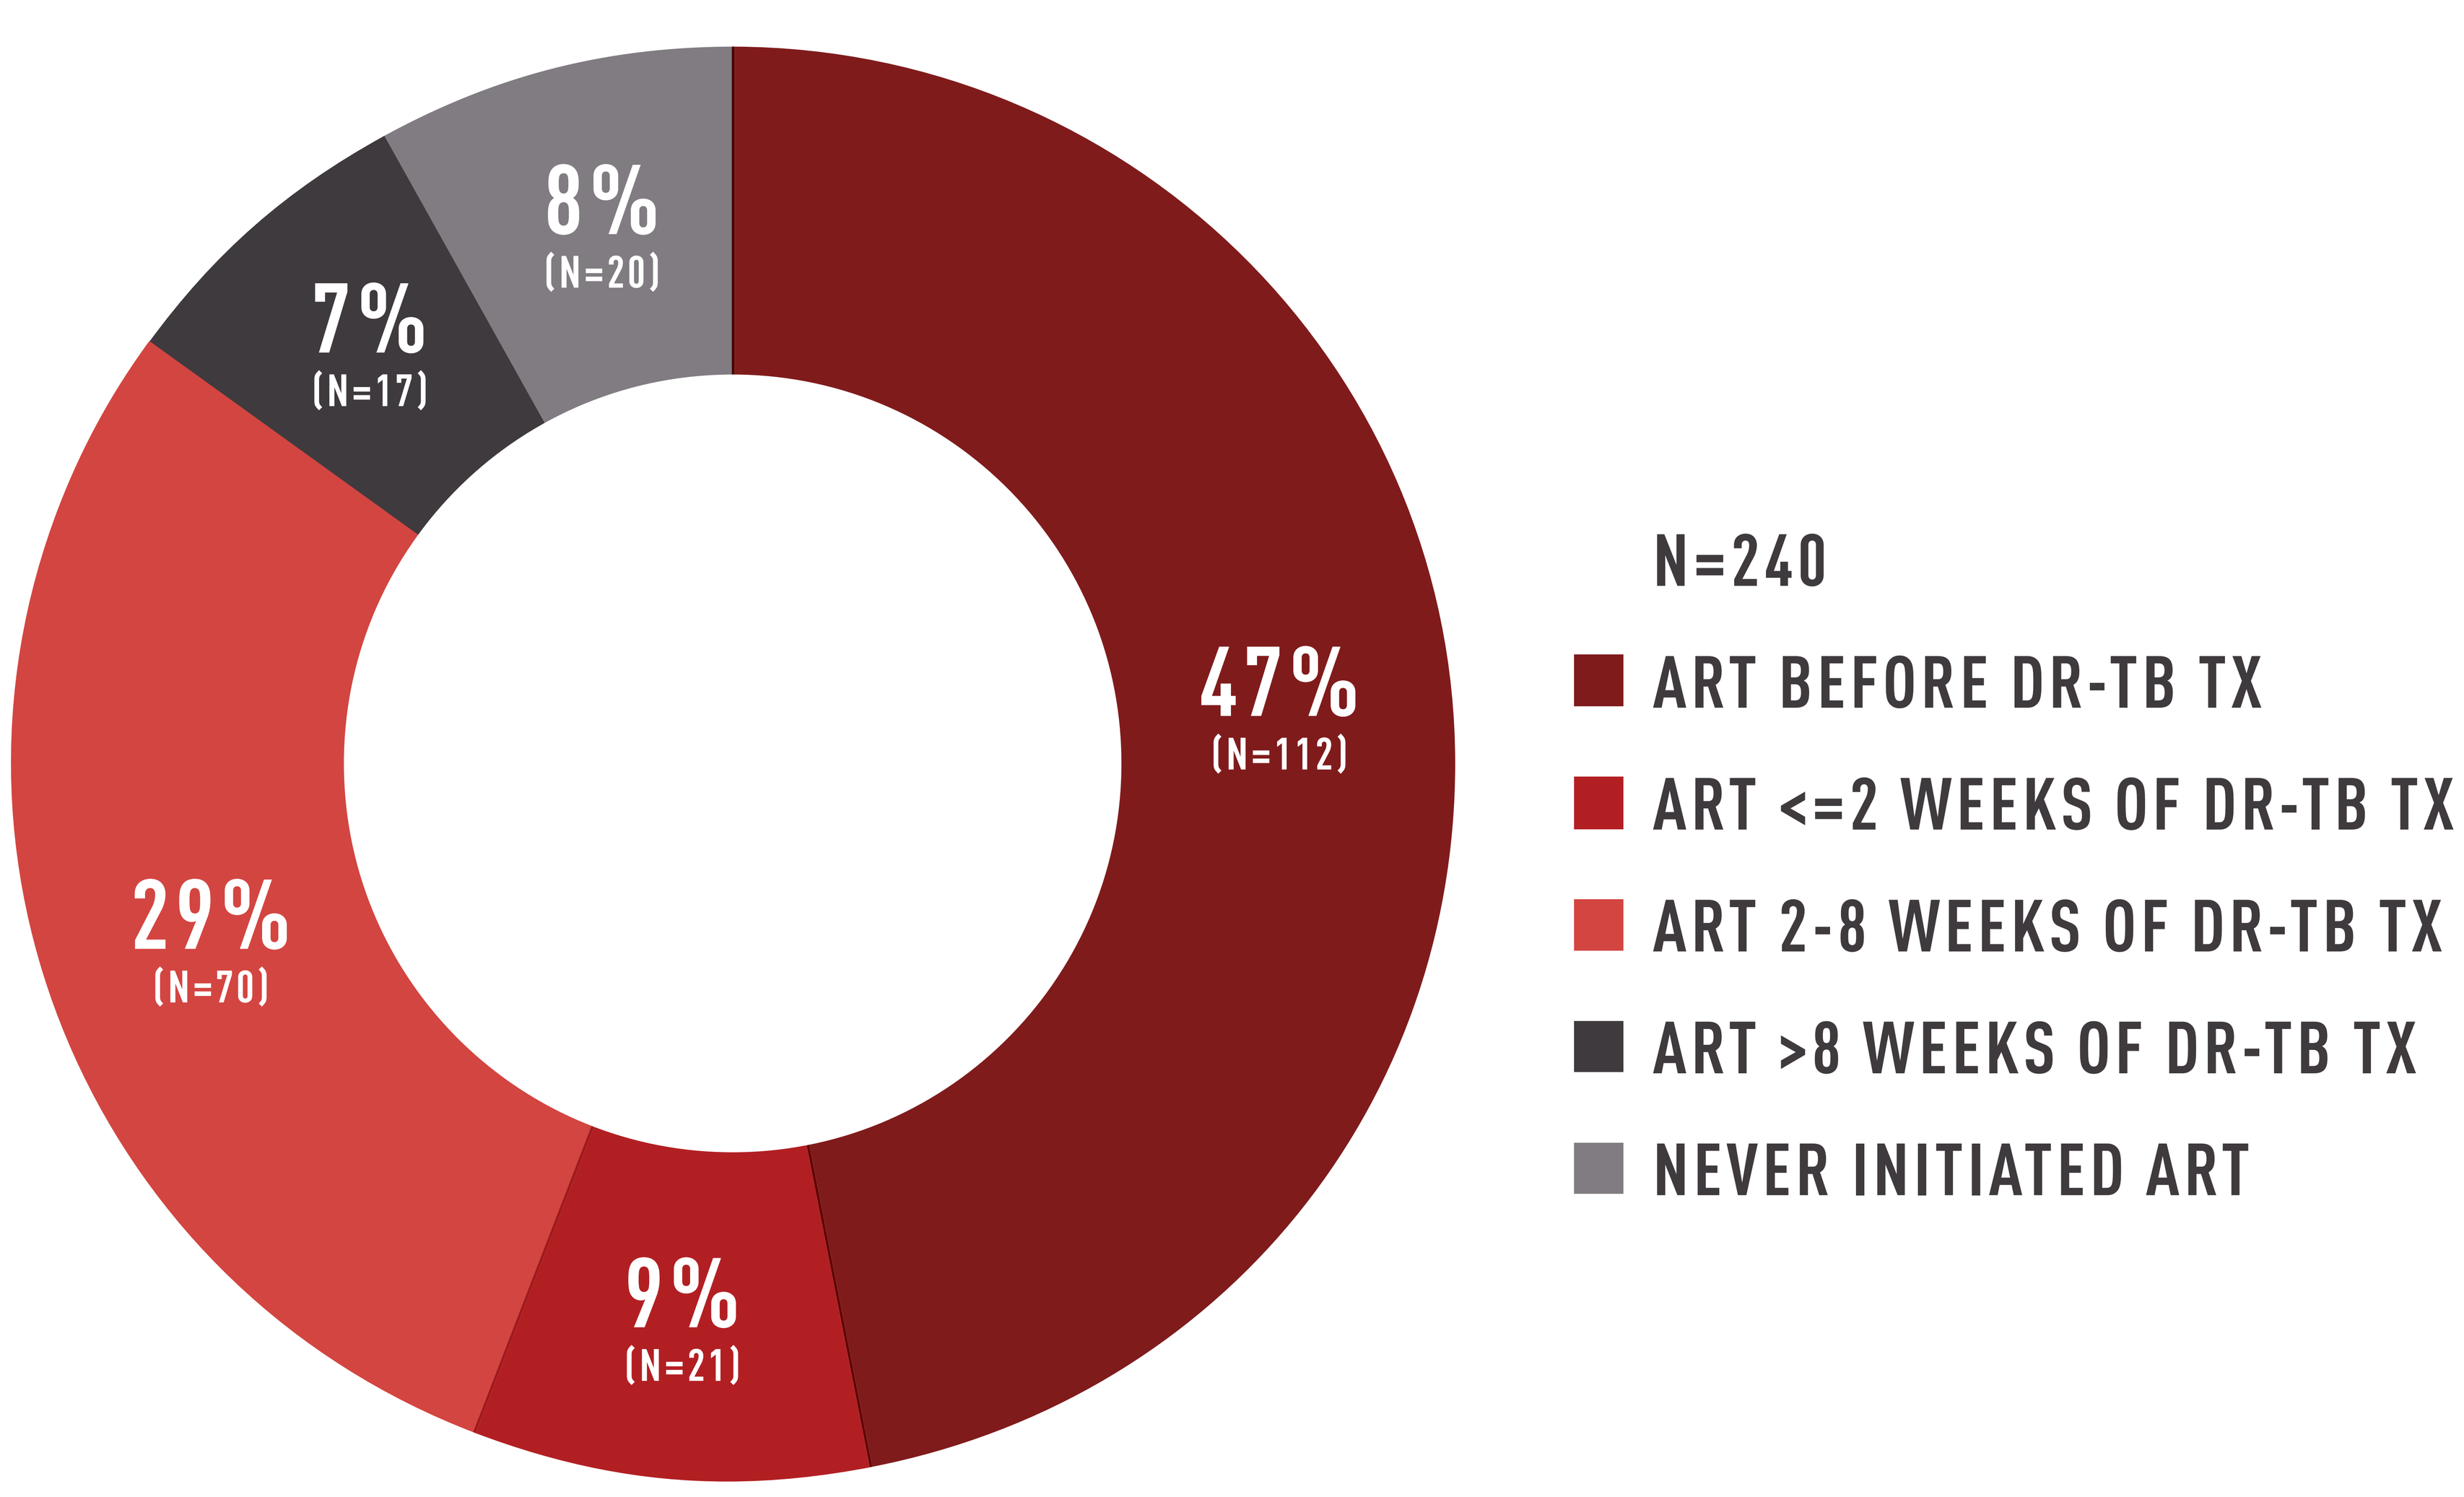
8%
(N=20)
7%
(N=17)
47%
(N=112)
29%
(N=70)
9%
(N=21)
N=240
ART BEFORE DR-TB TX
ART <=2 WEEKS OF DR-TB TX
ART 2-8 WEEKS OF DR-TB TX
ART >8 WEEKS OF DR-TB TX
NEVER INITIATED ART

Figure 1. *Time of ART initiation among those diagnosed with culture positive DR-TB from January 2014-December 2015*

RESULTS

Of 375 patients initiating treatment for pulmonary DR-TB, 286 (76.3%) were HIV-infected; 240/375 (64%) were culture-positive at treatment initiation, had available laboratory information and included. **Figure 1** displays time to ART initiation for patients included in the study. Among patients who initiated ART before DR-TB treatment median time on ART before DR-TB treatment was 8.4 weeks (IQR 1.6-28.2). Among patients who initiated ART following DR-TB treatment, median time to ART initiation was 3.9 weeks (IQR 2.4-5.2); median time to ART initiation if ART was started >8 weeks following DR-TB diagnosis, was 14.7 weeks (IQR 12.6-24.0). Mortality within 6-months of DR-TB treatment initiation was significantly higher among those that never initiated ART (8/20, 40.0%) compared to those that ever initiated ART (24/220, 10.9%) (OR 5.4, 95% CI 2.0-14.7, p=0.001) (**Figure 2**). Among patients still alive at 6-months, there were no significant differences in the proportions of patients with culture conversion if ART was initiated after DR-TB treatment (p=0.90). Additionally, those who never initiated ART had 4.3 times the odds (95% CI 1.3-14.2, p=0.016) of being culture positive at 6-months (7/12, 58.3%) compared to those who ever initiated ART (48/196, 24.5%); overall, 26.4% (55/208) of patients remained culture positive at 6-months.

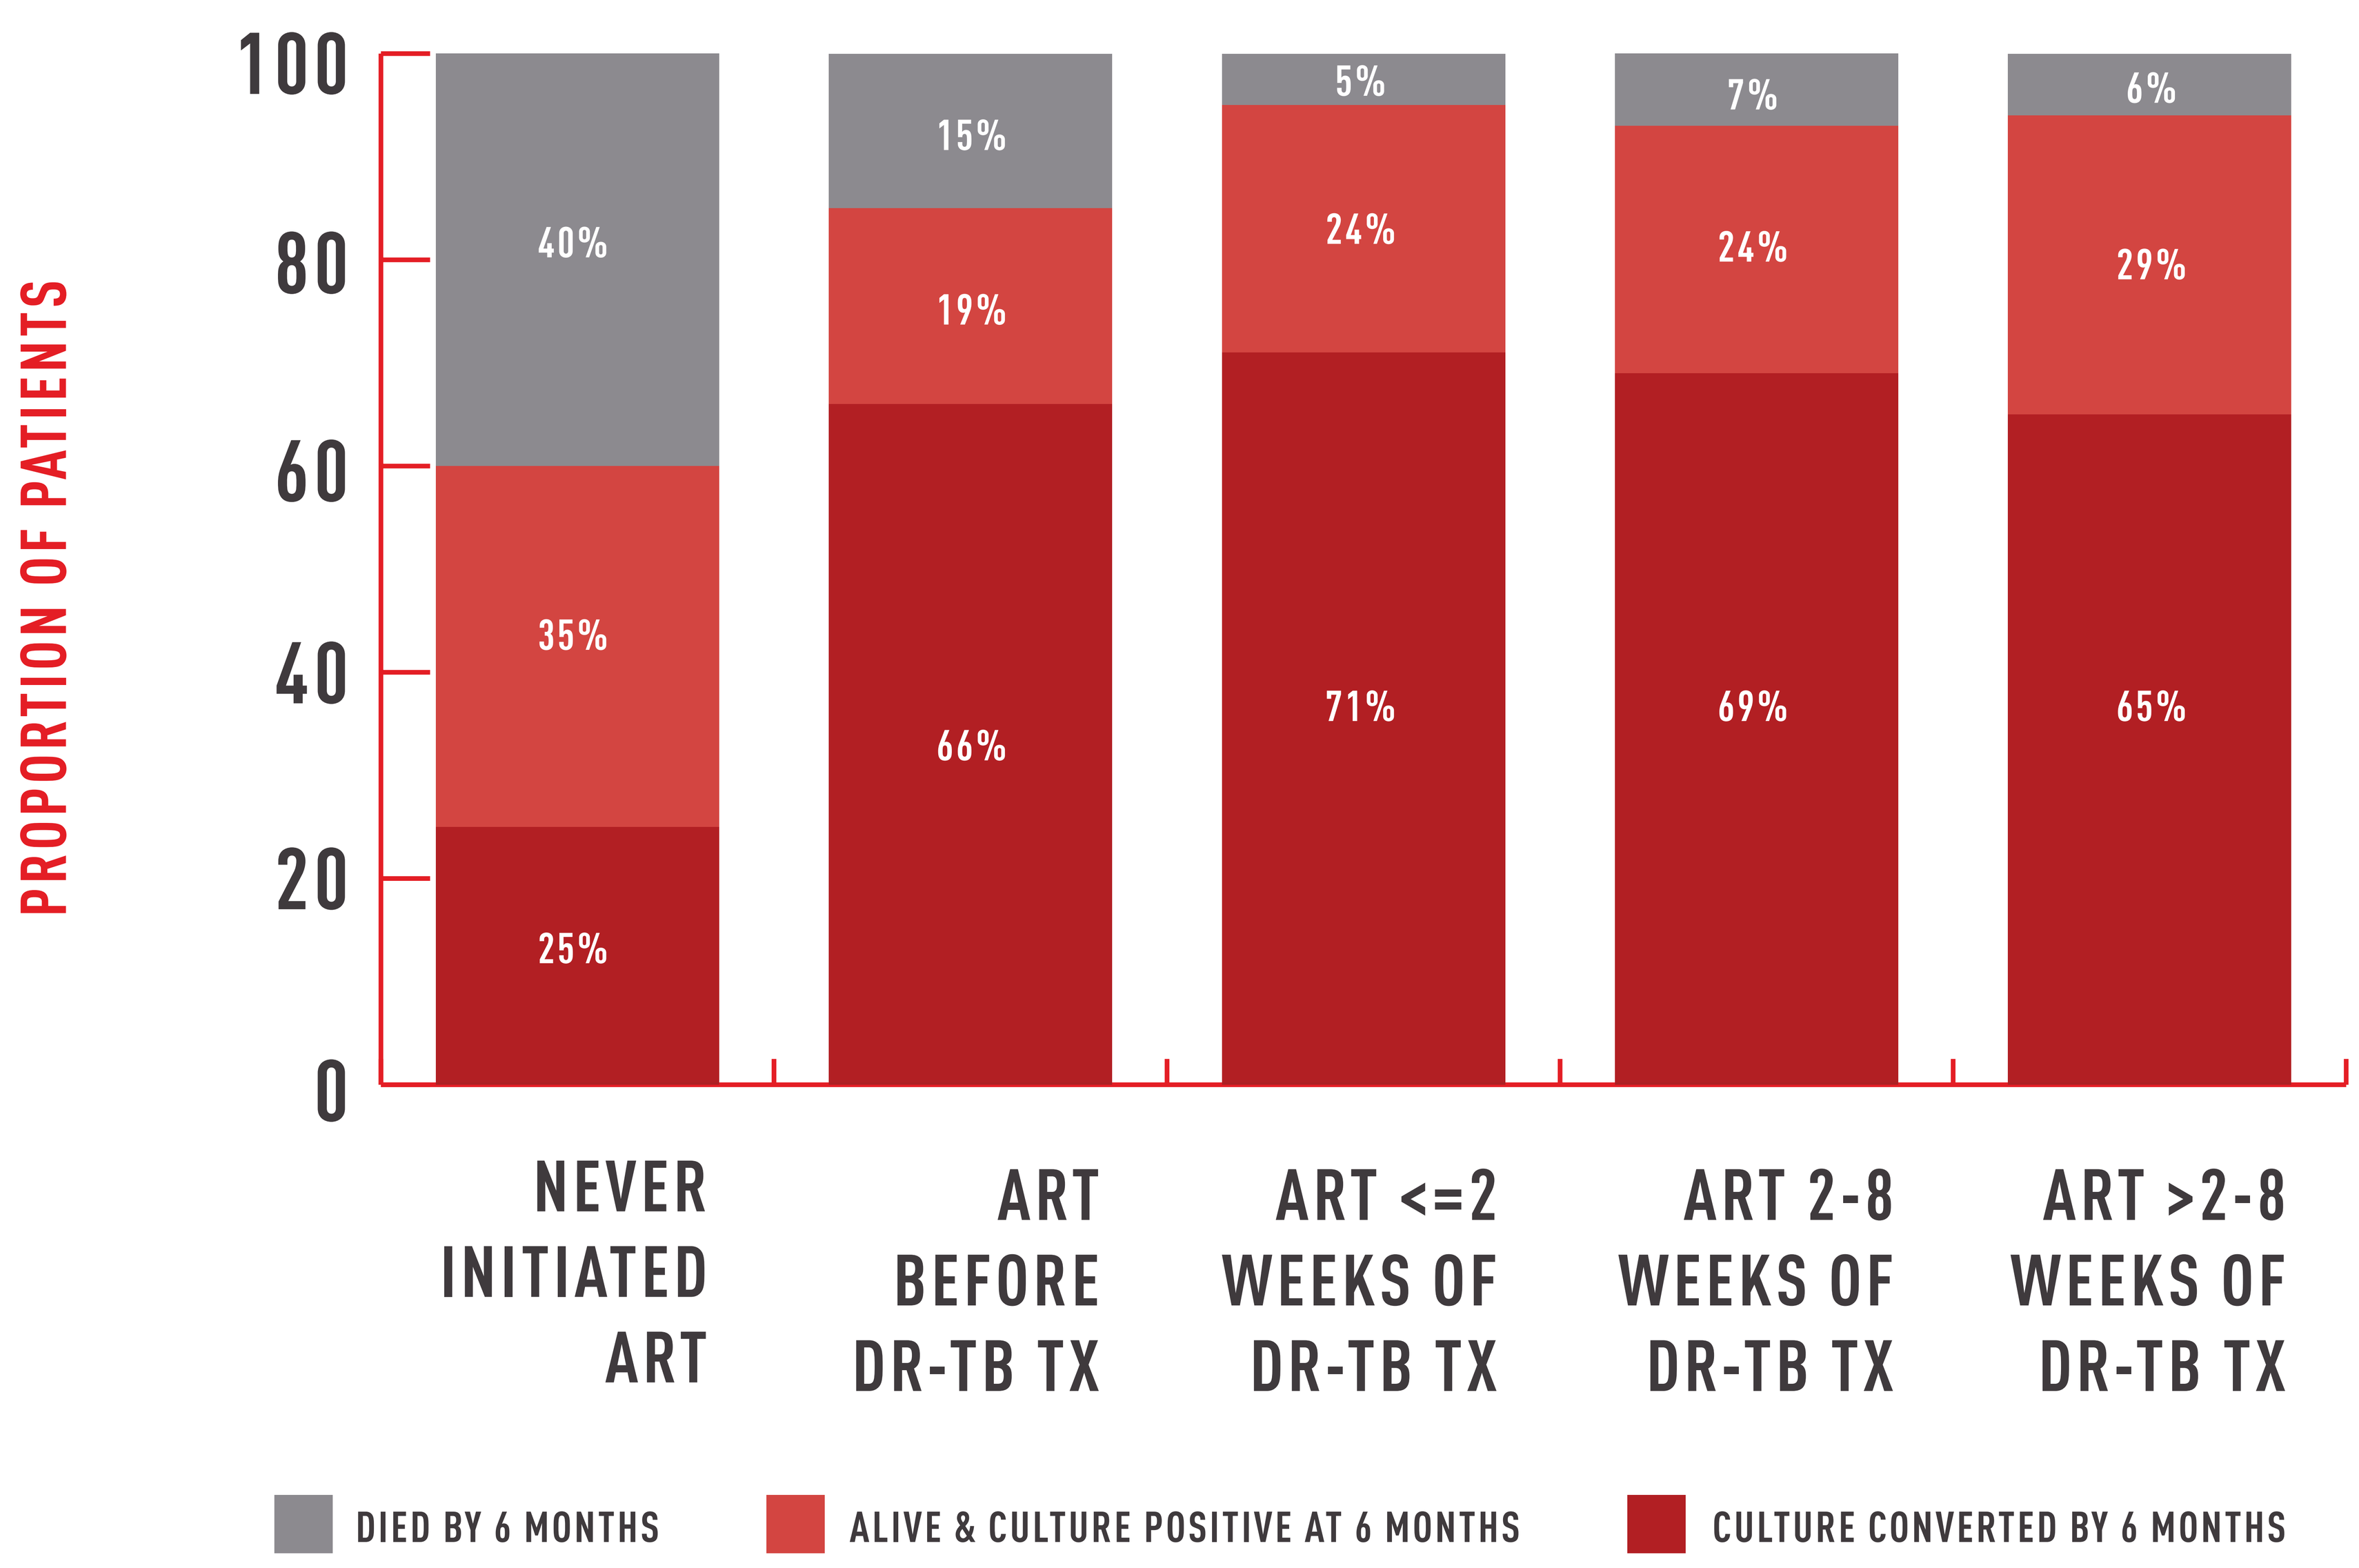
PROPORTION OF PATIENTS
100
80
60
40
20
0
40%
35%
25%
15%
19%
66%
5%
24%
71%
7%
24%
69%
6%
29%
65%
NEVER INITIATED ART
ART BEFORE DR-TB TX
ART <=2 WEEKS OF DR-TB TX
ART 2-8 WEEKS OF DR-TB TX
ART >2-8 WEEKS OF DR-TB TX
DIED BY 6 MONTHS
ALIVE & CULTURE POSITIVE AT 6 MONTHS
CULTURE CONVERTED BY 6 MONTHS

Figure 2. *Culture conversion following 6-months of DR-TB treatment, stratified by time to ART initiation*

CONCLUSIONS

Despite universal access to ART in South Africa, some DR-TB patients never receive ART increasing their risk of early morbidity and mortality. Clinical factors impacting time to culture conversion among those who initiate ART following DR-TB treatment need to be investigated in more detail. Over ¼ of the cohort was still culture positive following 6-months of DR-TB treatment; there is an urgent need for improved DR-TB regimens, additionally ART initiation among DR-TB patients should be prioritized.

ACKNOWLEDGEMENTS

- People suffering from DR-TB in Khayelitsha
- Western Cape Government Health & City of Cape Town Health Department
- National Department of Health
- Khayelitsha clinic staff